An Open-label Study of Fixed-Combination Halobetasol Propionate and Tazarotene Lotion for Psoriasis in Patients With Skin of Color

Neal Bhatia,[1] Andrew Alexis,[2] Seemal R. Desai,[3,4] Abby Jacobson[5]

[1]Therapeutics Clinical Research, San Diego, CA; [2]Weill Cornell Medical College, New York, NY; [3]Innovative Dermatology, Plano, TX; [4]The University of Texas Southwestern Medical Center, Dallas, TX; [5]Ortho Dermatologics (a division of Bausch Health US, LLC), Bridgewater, NJ

OBJECTIVE

- **To assess long-term efficacy and safety of once-daily halobetasol propionate (0.01%) and tazarotene (0.045%) lotion (HP/TAZ) in non-White and White participants with psoriasis**

CONCLUSIONS

- **With 52-week HP/TAZ treatment, both non-White and White participants with psoriasis experienced high rates of skin clearance, and comparable proportions experienced similar times to disease recurrence and reduced body surface area involvement**
- **Long-term treatment with HP/TAZ demonstrated improvements in local skin reactions in patients with psoriasis, with no new safety concerns**

INTRODUCTION

- Non-White patients may be more severely affected by psoriasis than White patients,[1] underscoring the need to examine topical therapies for psoriasis in this population
- The fixed-dose corticosteroid/retinoid combination halobetasol propionate (0.01%) and tazarotene (0.045%) lotion (HP/TAZ) is approved for topical treatment of plaque psoriasis in adults[2]
- Treatment with HP/TAZ has demonstrated treatment success (≥2-grade improvement from baseline in investigator's global assessment [IGA] and score of 0 [clear] or 1 [almost clear]) in non-White and White patients with psoriasis, with good tolerability and safety over 8 weeks[3]
- Here, we assessed the long-term efficacy and safety of once-daily HP/TAZ in non-White and White participants in a 52-week open-label study

METHODS

- This was a post hoc analysis of a phase 3, multicenter, open-label study of the long-term safety of HP/TAZ in participants with plaque psoriasis (NCT02462083)

Study design

- Participants (non-White, n=77; White, n=473) aged ≥18 years with IGA of 3 or 4 and body surface area (BSA) involvement between 3% and 12% were included
- Once-daily HP/TAZ was administered for 8 weeks
- Treatment success (IGA of clear [0] or almost clear [1]) or treatment failure (IGA ≥2) was used to determine the need for retreatment after the initial 8-week course and subsequent 4-week courses
- At week 8, participants who achieved the primary endpoint of treatment success stopped treatment with HP/TAZ and were reevaluated at week 12; those with treatment failure at week 8 received once-daily HP/TAZ for an additional 4 weeks
- At week 12, participants with ≥1-grade improvement in IGA from baseline continued the study
- Participants were allowed up to 24 continuous weeks of HP/TAZ treatment and were reevaluated monthly for achievement of IGA 0/1
- Subsequent decisions to continue or discontinue treatment at each monthly evaluation were repeated for up to 1 year
 - If at any point IGA ≥2 was achieved, HP/TAZ was resumed, and if IGA 0/1 was achieved, HP/TAZ was discontinued

Data analysis

- Non-White and White participants who completed up to 1 year of the study were included in this post hoc analysis
- Long-term efficacy of HP/TAZ was assessed using treatment success, time to disease recurrence, and BSA involvement
 - Time to retreatment was analyzed in participants who stopped HP/TAZ after achieving treatment success at or after week 8
 - BSA involvement was assessed in participants who remained in the study for 1 year
- Safety and tolerability were evaluated through assessment of selected local signs and symptoms (itching, dryness, and burning/stinging)

RESULTS

Long-term efficacy

- At week 52, 33.3% of non-White and 40.3% of White participants achieved treatment success
- The proportion of participants who did not experience disease recurrence was similar between the non-White and White groups (~7%)
- In participants who achieved treatment success on or after week 8 and stopped HP/TAZ, the proportion of participants who did not require retreatment (ie, first postcessation evaluation in which treatment success was not achieved) was similar between the non-White and White groups after cessation of HP/TAZ (Figure 1)

Figure 1. Non-White and White participants who did not require retreatment after first treatment success with HP/TAZ on or after week 8.

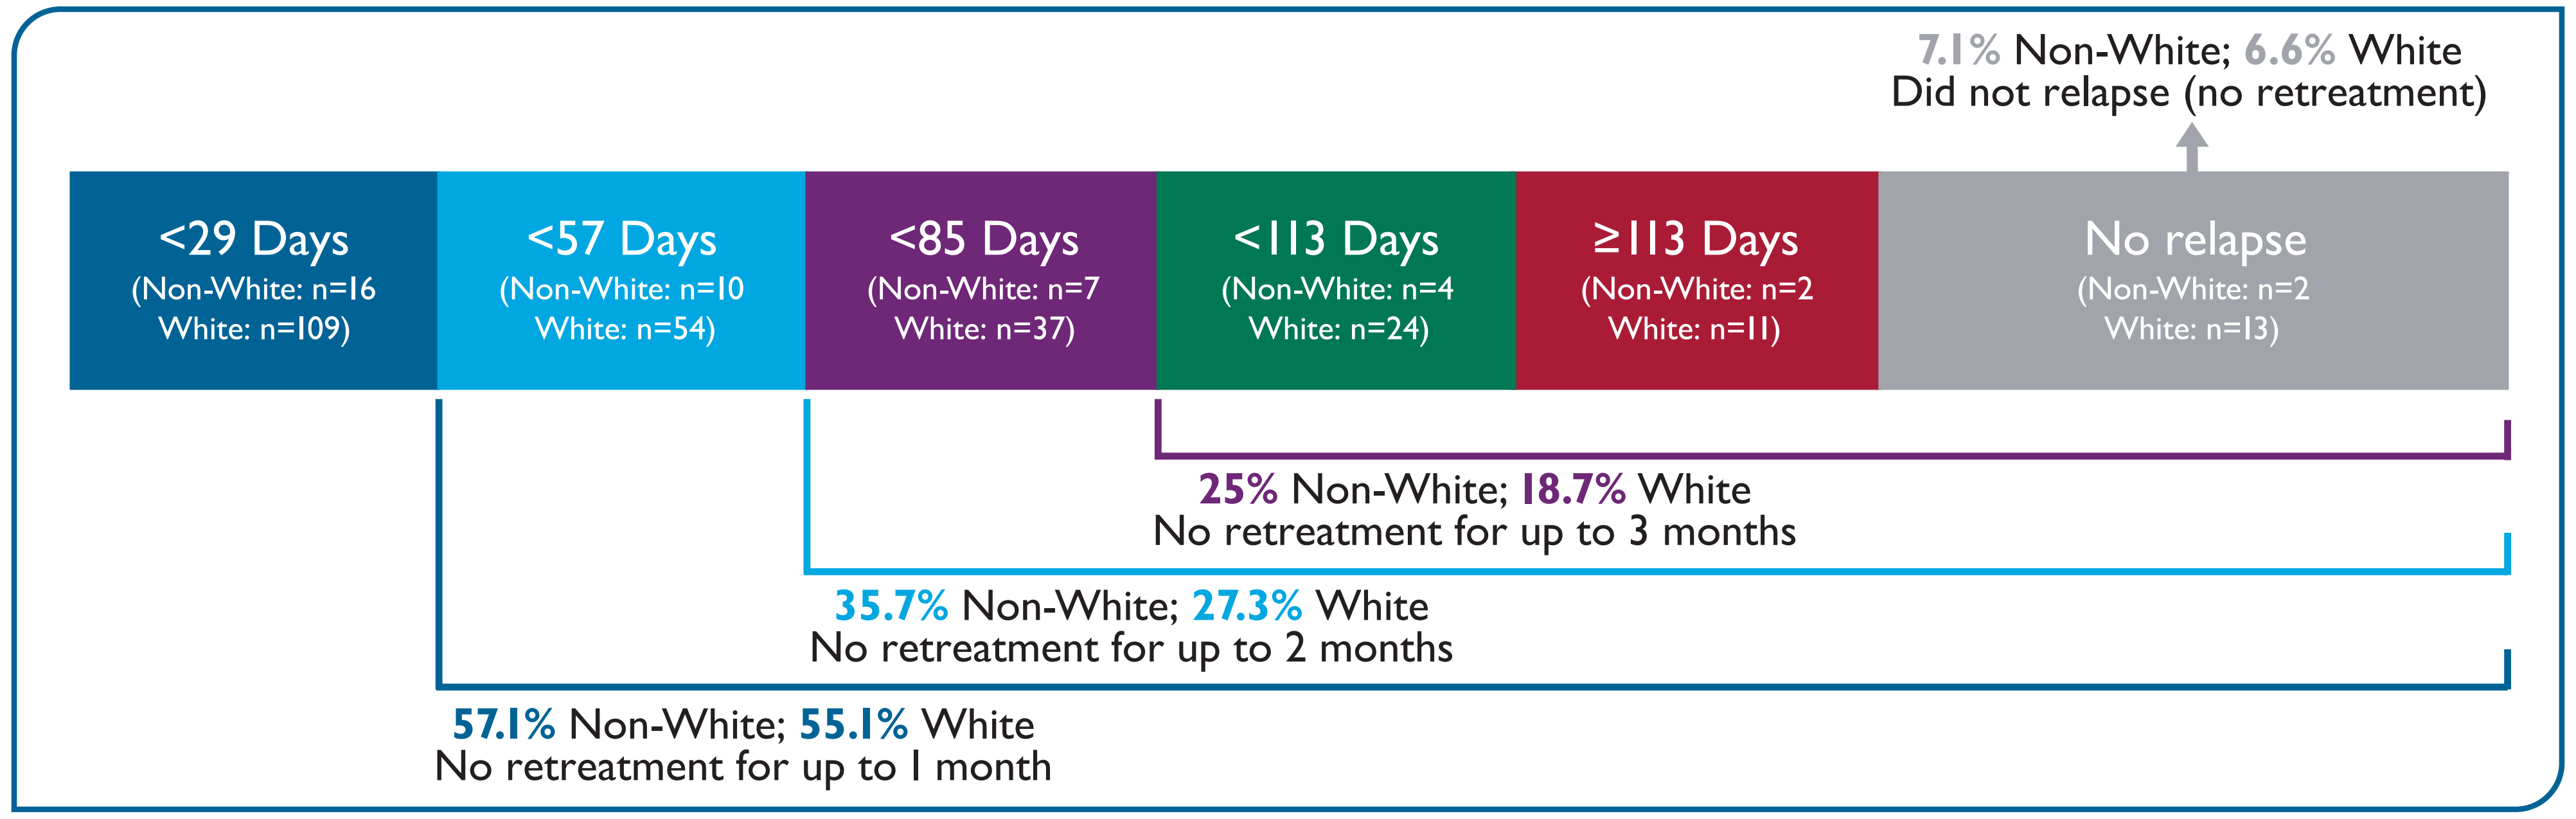
7.1% Non-White; 6.6% White
Did not relapse (no retreatment)
<29 Days (Non-White: n=16 White: n=109)
<57 Days (Non-White: n=10 White: n=54)
<85 Days (Non-White: n=7 White: n=37)
<113 Days (Non-White: n=4 White: n=24)
≥113 Days (Non-White: n=2 White: n=11)
No relapse (Non-White: n=2 White: n=13)
25% Non-White; 18.7% White
No retreatment for up to 3 months
35.7% Non-White; 27.3% White
No retreatment for up to 2 months
57.1% Non-White; 55.1% White
No retreatment for up to 1 month

HP/TAZ, halobetasol propionate (0.01%) and tazarotene (0.045%) lotion.

- Among individuals participating in the study for at least 1 year, the proportion of non-White and White participants who maintained BSA involvement ≤3% from week 8 to end of study was comparable (Figure 2)

Long-term safety and tolerability

- At baseline, the proportion of participants with no itching, dryness, or burning/stinging was comparable between non-White and White participants (33.8% vs 23.7%; 37.7% vs 31.3%; 68.8% vs 66.6%, respectively)
- At week 52, a numerically lower proportion of non-White participants had no itching or dryness compared with White participants (Figure 3)
 - >88% of participants in each group did not experience any burning/stinging at week 52

Figure 2. Participants who remained in the study for at least 1 year who maintained BSA involvement ≤3% from week 8 to end of study.

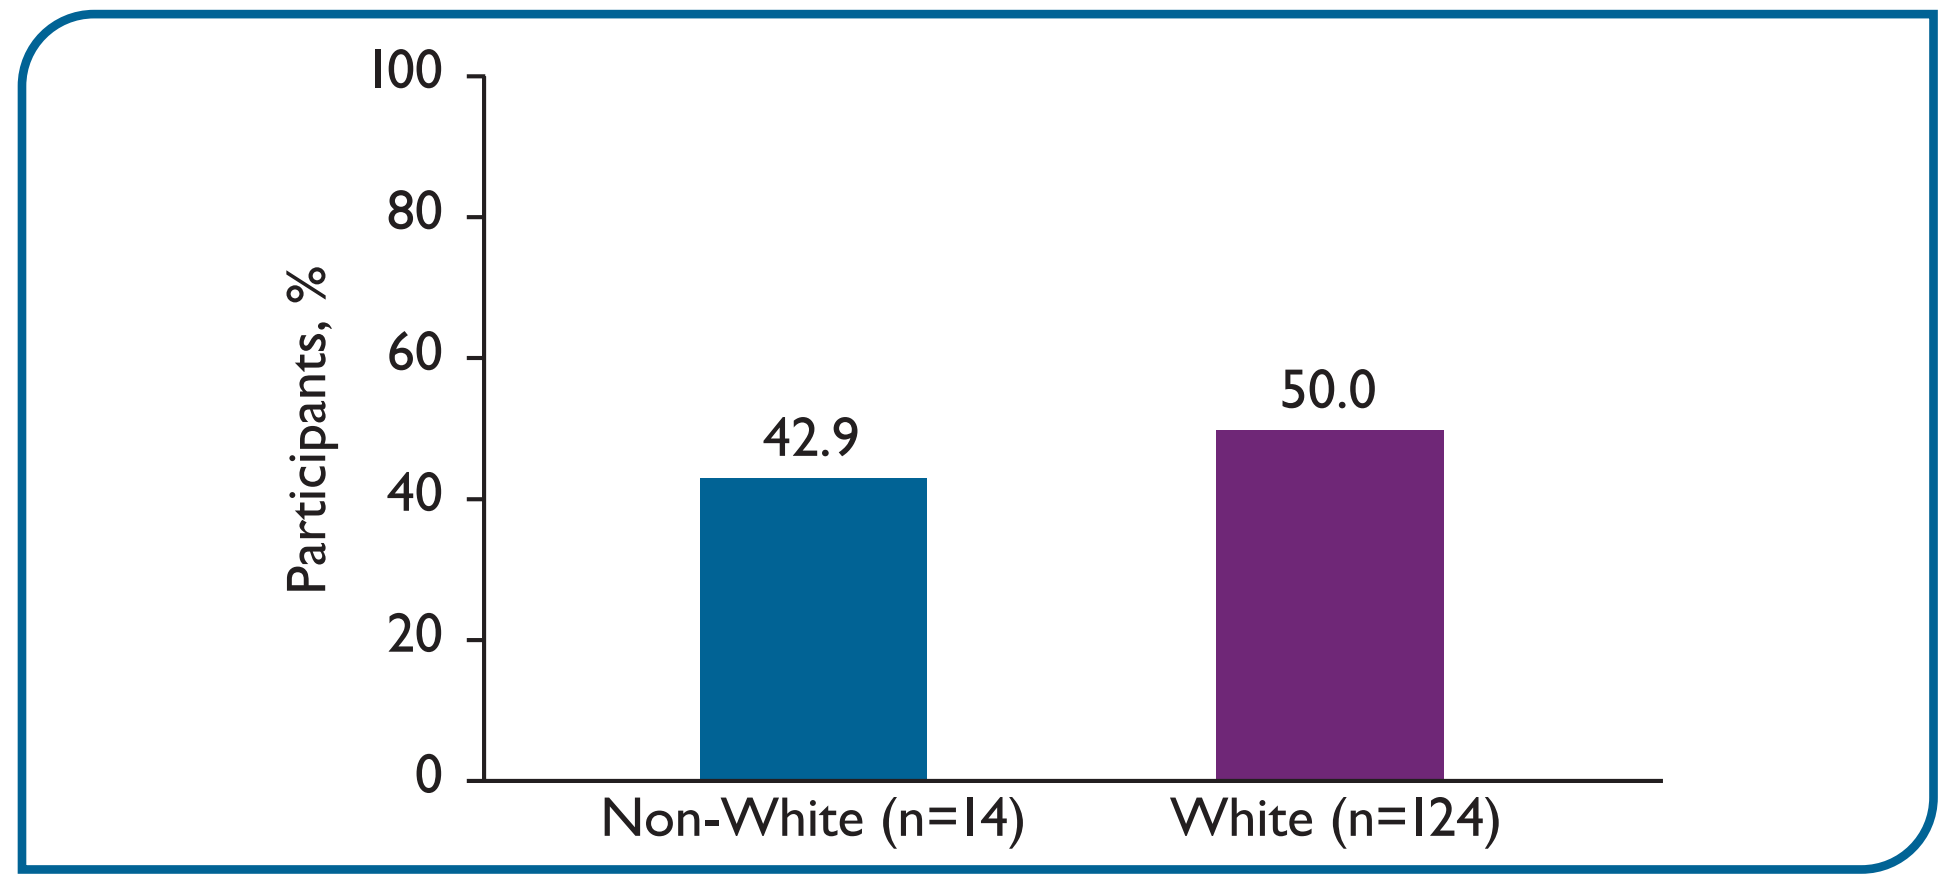
100
80
60
40
20
0
Participants, %
42.9
50.0
Non-White (n=14)
White (n=124)

Analysis was performed based on maximum day related to visits or participant disposition (end of study), and whether that exceeded 358 days, and may not directly coincide with attendance at the week 52 visit. BSA, body surface area.

- Throughout the 52-week study, rates of adverse events were similar between non-White and White participants (53.2% vs 57.7%, respectively)
 - Few participants (non-White, 5.2%; White, 7.8%) discontinued HP/TAZ because of adverse events
- Application site reaction was the most common treatment-related adverse event for both non-White (26.0%) and White (31.5%) participants

Figure 3. Percentage of non-White and White participants with no local skin reactions at week 52.

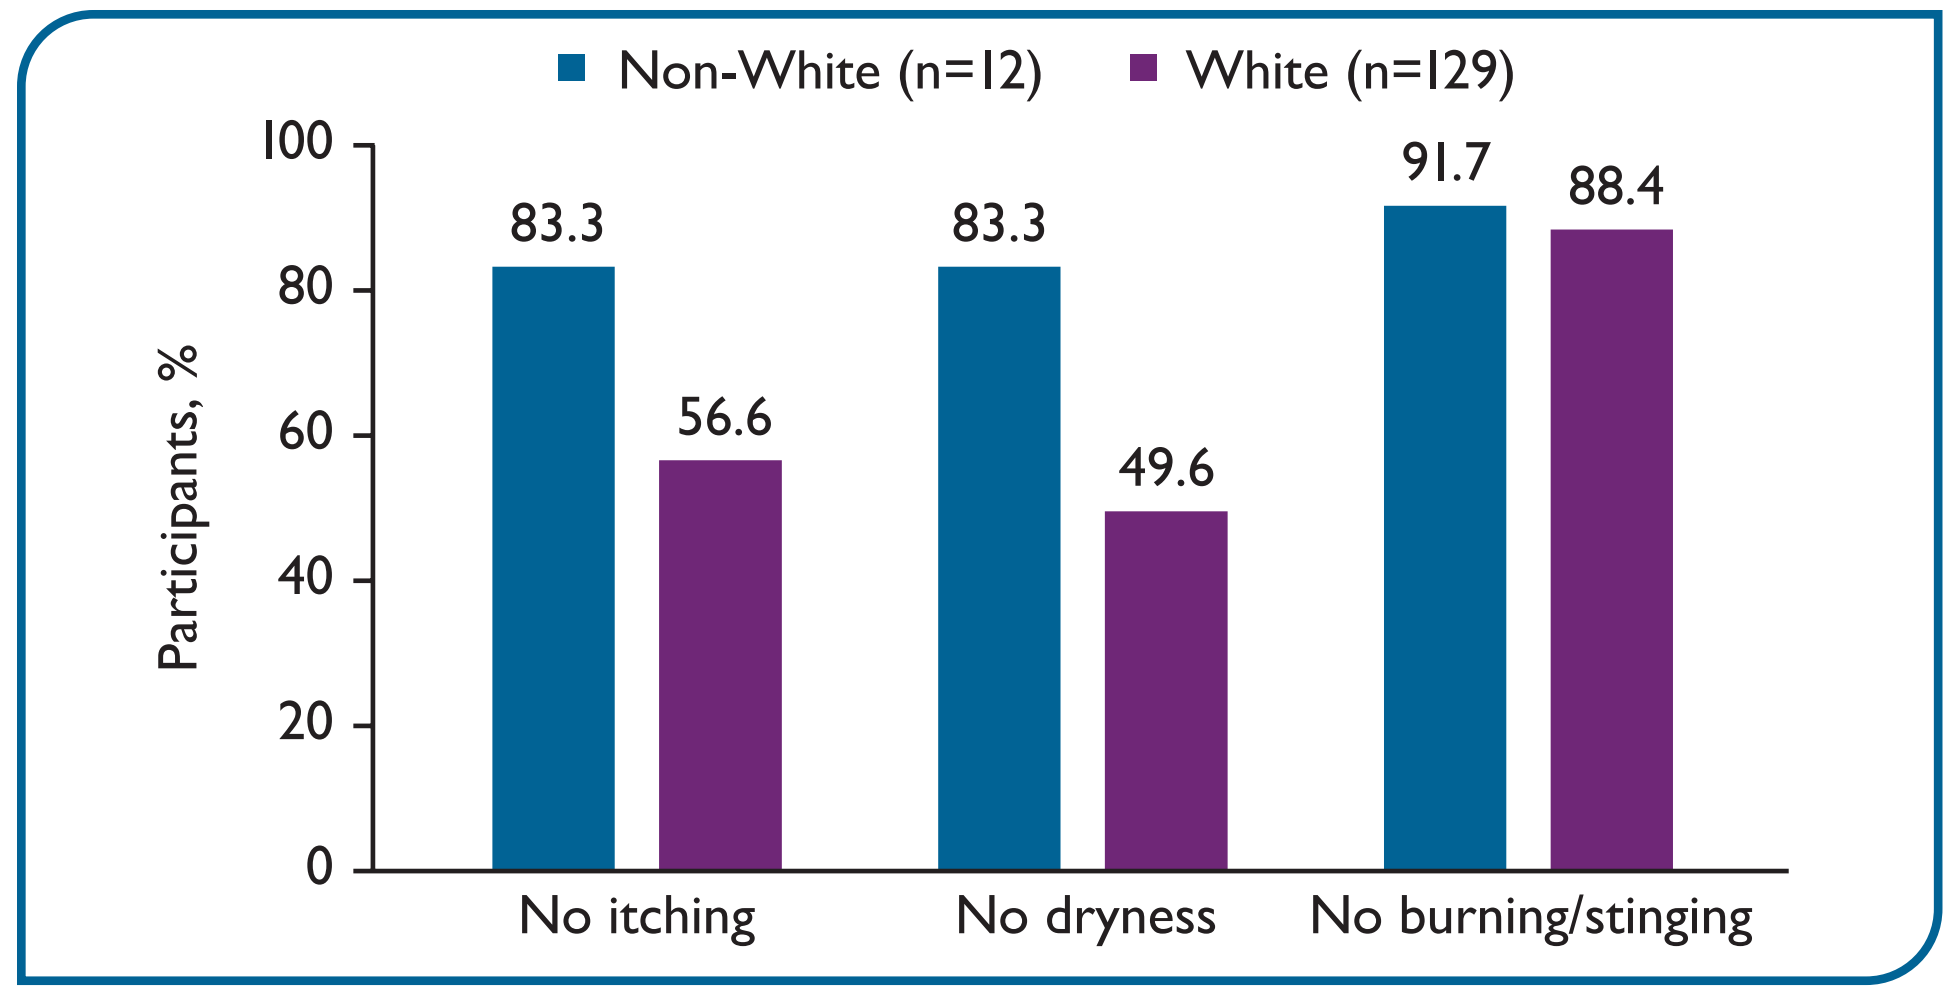
Non-White (n=12)
White (n=129)
100
80
60
40
20
0
Participants, %
83.3
56.6
83.3
49.6
91.7
88.4
No itching
No dryness
No burning/stinging

Acknowledgments: This study was sponsored by Ortho Dermatologics. Medical writing and editorial support were provided by MedThink SciCom and funded by Ortho Dermatologics. Ortho Dermatologics is a division of Bausch Health US, LLC.

References: **1.** Alexis and Blackcloud. *J Clin Aesthet Dermatol.* 2014;7:16-24. **2.** DUOBRII [package insert]. Bausch Health US, LLC; 2020. **3.** Alexis et al. *J Drugs Dermatol.* 2021;20:744.

Skin of Color Update • September 10-12, 2021 • Virtual